cHSP60-IGG-ELISA MEDAC: EVALUATION OF A NEW RESEARCH ASSAY FOR DETECTION OF IGG ANTIBODIES TO CHLAMYDIAL HEAT SHOCK PROTEIN 60

Dreesbach K[1], König T[1], Christiansen G[2], Henriksen T[2], Pedersen AS[3], Birkelund S[3], and Franke D[1]

[1]medac GmbH, Hamburg, Germany, [2]University of Aarhus, Denmark, and [3]Loke Diagnostics ApS, Aarhus, Denmark

ABSTRACT

We evaluated the new cHSP60-IgG-ELISA medac with regard to precision, suitability for automation and quantitation. cHSP60 IgG antibody prevalence in different cohorts was also determined.

The validation data which we have obtained demonstrated a good reproducibility in terms of intra-assay and interassay variance. Moreover, a good correlation between automatically and manually performed test runs was found. Finally, reliable end-titers can be calculated from a single OD measurement.

The comparison of antibody prevalence in small numbers of blood donors and children with various small patient groups revealed partly distinct differences. Elevated antibody prevalence and more sera with high titers were observed in both the infertility group and in patients with reactive arthritis.

INTRODUCTION

Heat shock proteins are highly conserved proteins which are expressed in both procaryotes and eucaryotes with pronounced homology. Chlamydial heat shock protein 60 (cHSP60) shares an amino acid identity with its homolog in other bacteria of approximately 60%, of about 50% with its human homolog, and an identity of > 95% within the genus chlamydia.

Chlamydial heat shock protein 60 (cHSP60) has been suspected to play a role in immunopathogenesis of chlamydial disease. Moreover, an increasing number of investigations has shown high prevalence of cHSP60 serum antibodies among patients with sequelae of chlamydial infections like pelvic inflammatory disease (PID), adverse outcome of pregnancy, ectopic pregnancy (EP), tubal factor infertility (TFI), *in vitro* fertilization (IVF) failure, and reactive arthritis.

To date, however, the diagnostic value of cHSP60-specific serum antibodies has been discussed with controversy. A commercial assay suited for routine use should improve the comparability of results obtained in different laboratories.

METHODS

The cHSP60-IgG-ELISA medac is an indirect ELISA using recombinant full-length HSP60 from *Chlamydia trachomatis*. The sequence of the cloned gene was shown to be identical with published data. The cHSP60 was produced as a hexahistidine-tagged protein in *E. coli*.

For cut-off determination 44 sera without chlamydial IgG antibodies were measured. The optical density (OD) median plus threefold standard deviation (SD), excluding outliers, was defined as the cut-off value. The grey zone was set to cut-off ± 10%.

For the calculation of precision data six sera of different reactivity were used for intra-assay variance (performing a 22-fold determination of each sample), and seven sera for interassay variance (performing 11 independent test runs).

The accuracy of end-titer calculation with the formula provided in the package insert was evaluated by titration of twelve sera.

Automation suitability was investigated using the Dynatech Immuno-Assay System (DIAS). In parallel to manually performed test runs the same samples for intra-assay (10-fold determination) and interassay variance (11 independent test runs) as well as 31 different sera from the cohorts mentioned below were measured on the DIAS. Furthermore, 12 sera were tested manually and on the Behring ELISA Processor (BEP) III in parallel.

Serum antibody prevalence was determined in the following cohorts: 100 volunteer blood donors, 37 children (age 0-17 years), 68 female infertility patients (secondary sterility or laparoscopically diagnosed TFI), and 44 patients with reactive arthritis.

RESULTS

In two different kit lots the OD median plus 3 SD of the negative sera was determined as 0.368 and 0.352, respectively. The cut-off was set to 0.35 plus OD of the negative control contained in each kit.

Coefficients of variation (CV) from sera within the relevant OD range (positive or equivocal) were found below 10.5% for both manually and automatically performed investigations.

Fig. 1 shows the correlation of calculated end-titers and the corresponding titration results (expected end-titer). Omitting high OD values between 2 and 3 resulted in slightly better correlation parameters (solid line). The difference between calculated and expected end-titer was within ± one dilution step.

The correlations of test results obtained manually and using both automated devices are shown in Fig. 2. Out of the 31 sera measured manually and on the DIAS, five were consistently outside the measuring range (OD > 3). No discrepant results were observed either with the DIAS or with the BEP III.

The following cHSP60 IgG prevalence was found:

- Volunteer blood donors 14%
- Children 8%
- Infertility patients 60%
- Patients with reactive arthritis 64%

The measured OD/cut-off values in each cohort are plotted in Fig. 3. The groups of infertility and reactive arthritis patients contain considerably more high positive samples than the other ones.

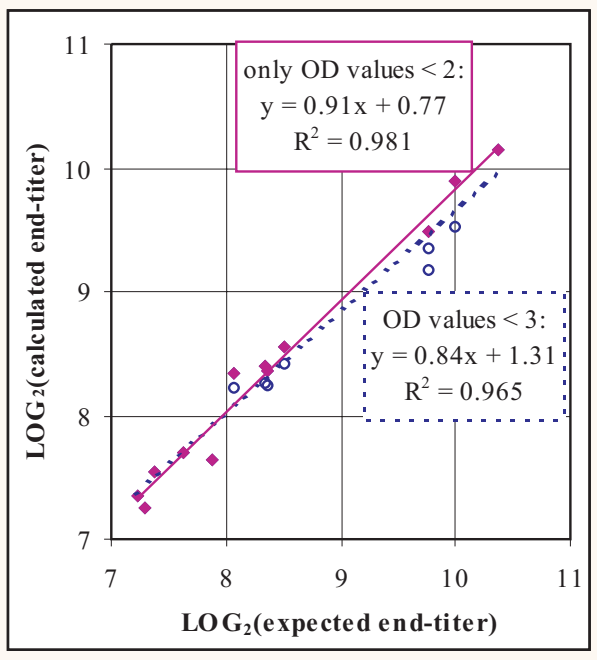
only OD values < 2:
y = 0.91x + 0.77
R^2 = 0.981
OD values < 3:
y = 0.84x + 1.31
R^2 = 0.965
LOG_2(calculated end-titer)
LOG_2(expected end-titer)
7
8
9
10
11

Fig. 1: Quantitation
Correlation of calculated versus expected (extrapolated from the last positive or borderline dilution) end-titer. All titers were not rounded to the next whole titration step. (Open circles: OD values between 2 and 3.)

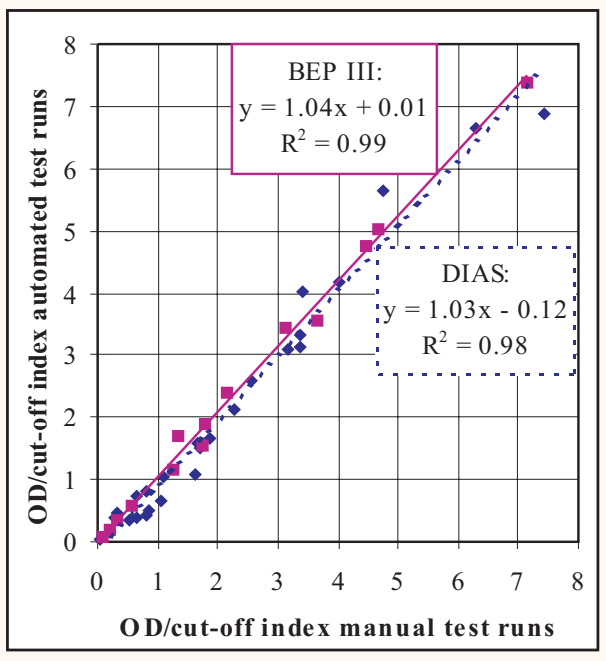
BEP III:
y = 1.04x + 0.01
R^2 = 0.99
DIAS:
y = 1.03x - 0.12
R^2 = 0.98
OD/cut-off index automated test runs
OD/cut-off index manual test runs
0
1
2
3
4
5
6
7
8

Fig. 2: Automation suitability
Correlation of OD/cut-off indices obtained with manually performed and automated test runs.

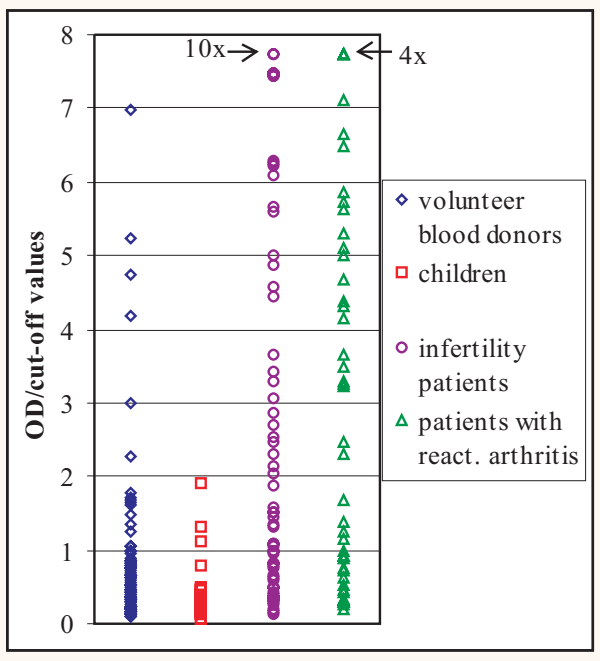
10x
4x
OD/cut-off values
volunteer blood donors
children
infertility patients
patients with react. arthritis
0
1
2
3
4
5
6
7
8

Fig. 3: Scatter plot of all measured OD/cut-off values in each cohort
OD values outside the measuring range were set to 3. The number of these samples are indicated besides the arrows.

CONCLUSIONS

Concerning the technical performance, the commercialized cHSP60-IgG-ELISA medac is suitable for routine testing and fulfills the demand for comparable results from various laboratories in the future..

Our first cHSP60 IgG prevalence data, particularly in patients with sequelae due to chlamydial upper genital tract infections, seem to be promising concerning the diagnostic usefulness of this assay. Further studies in larger cohorts have to be performed in order to work out the diagnostic value of cHSP60 antibody determination in such diseases.